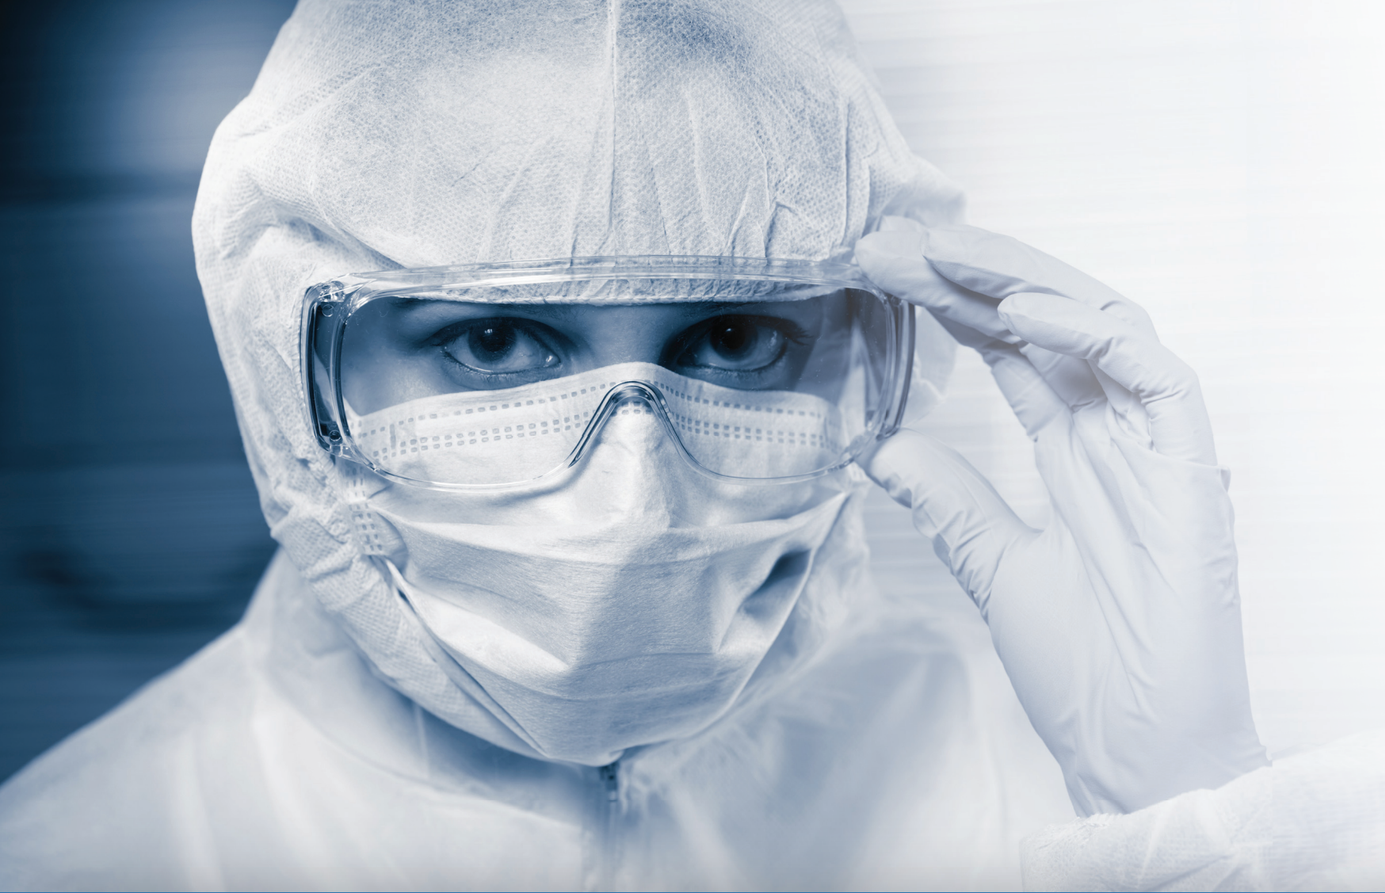

Practical Applications and Benefits of Sterile Product Compliance Risk Assessments & Case Studies

DR. GÜNTHER GAPP
GAPP QUALITY GMBH
6230 BRIXLEGG, AUSTRIA
www.gappquality.com

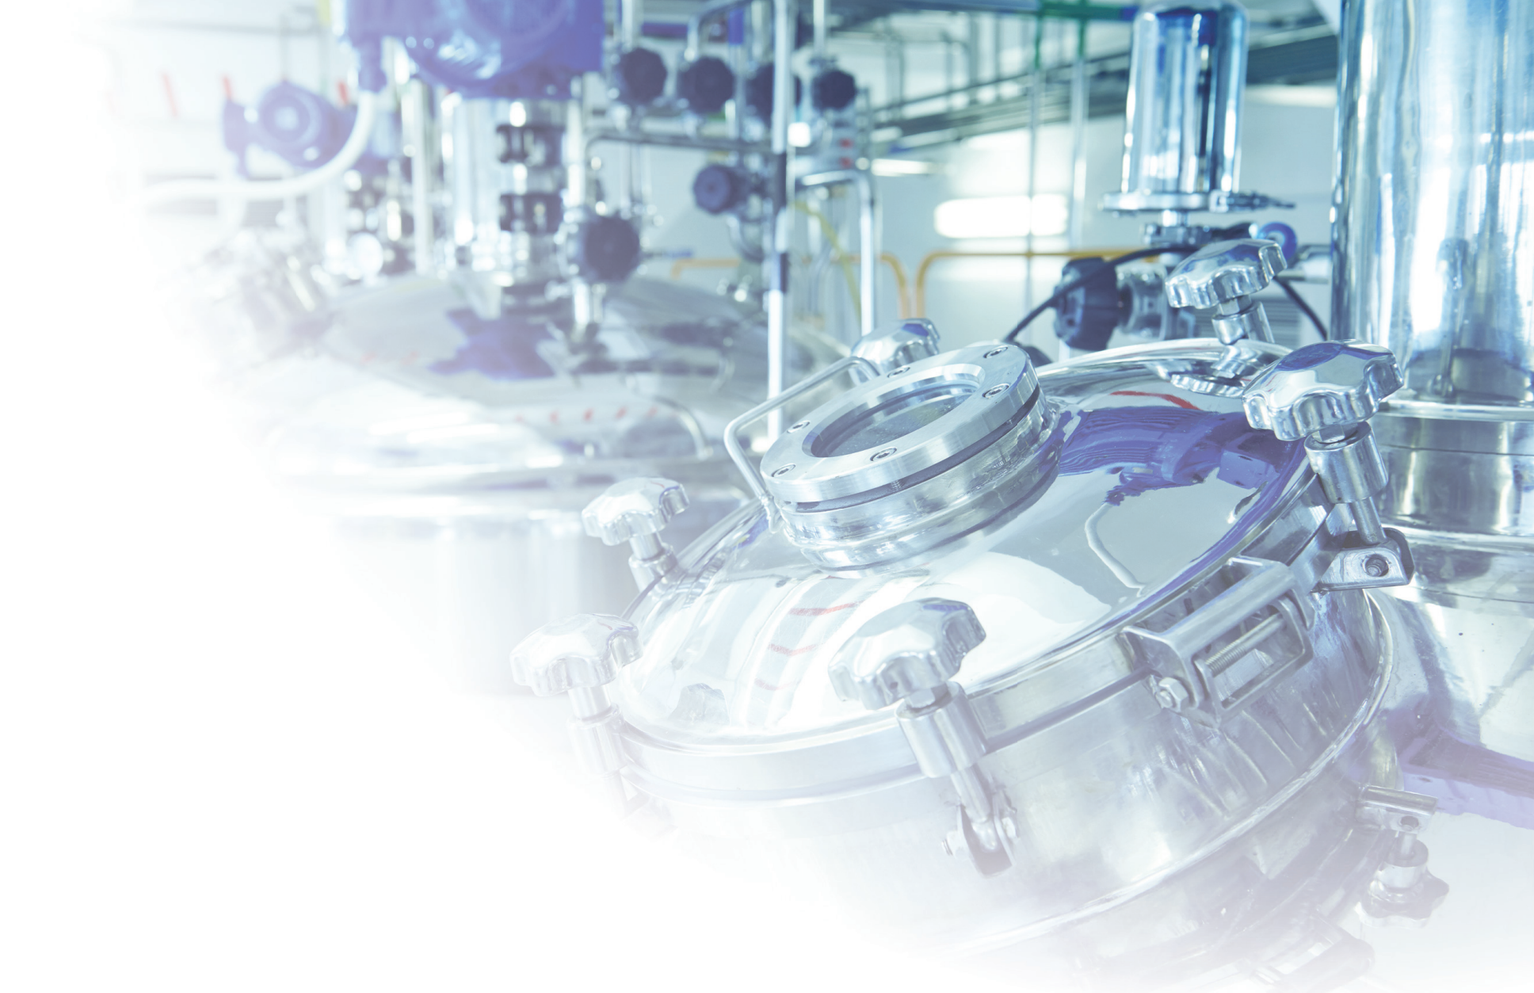

Reality Check & Responsibility

- Microorganisms are omnipresent in the environment
- A variety of potential root causes can lead to non-sterile results & product
- The whole batch must meet the sterility and endotoxins quality specifications for sterile products
- Neglecting appropriate precautions creates very high risk for the patient

- It is imperative to understand all possible microbial entry points and to implement quality risk management (QRM) strategies that aim to prevent microbial contamination.
- A systematic approach is highly recommended both, when developing proactive microbial control strategies or when investigating microbial contamination deviations.

Risk Assessments Are Required

Each sterile manufacturing production site is required to have a risk analysis in use [1]

- Comply with FDA 2004 initiative – Pharmaceutical cGMPs for the 21st century [1]
- Support the implementation of ICH Q9 and Q10 [2,3]
- Adhere to ISO 14971 [4]
- Mandatory risk analysis: The EU Good Manufacturing Practice (GMP) Guidelines [5]

Risk Analysis Tools

- Uncover potentially hazardous process steps and lacking controls
- Provide information about potential microbial contamination
- Are investigative in case of deviations for root cause analysis and batch disposition decisions
- Proactively help prevent non-compliance during product quality and regulatory audits

Gapp Quality SPCRA Tool – Facts

- Based on more than 20 year experience in the pharmaceutical industry [6]
- Simple, feasible and comprehensive approach to the risk analysis of sterile plants.
- Periodically revised tool comprising 243 specific questions (version 2017 vs. 203 questions version 2015 for FDF with sterile filtration)
- Relevant to all aseptic processing activities, comprising 243 specific questions that cover (e.g.):

Sterile filtration | Housekeeping | Cleaning and disinfection programs
(Re)Qualification of operators | Media fills | Environmental monitoring and trending
Isolator and restricted access barrier systems (RABS) equipment
Sterility testing | Holding time studies | Packaging integrity

- Individualized approach for active product ingredient (API) and finished dosage form (FDF) plants (with or without sterile filtration).
- Uncovers potential weaknesses of the process
- Enables proactive corrective and preventive actions (CAPAs) for further systematic improvement.

Gapp Quality SPCRA Tool – Method

The manufacturing steps are classified into individual **UNITS** according to their process flow. For each UNIT, a multitude of specific questions are asked, encompassing all areas of risk involved in aseptic processing. [6]

The following production UNITS are analyzed using Hazard Operability (HAZOP) analysis risk analysis method (for a FDF plant):

A **RISK EMPHASIS FACTOR (REF)** is introduced reflecting the inherent risk of the underlying UNIT on the overall sterility of the final product. The REF can take on a value of 1 (low), 2 (relatively low), 3 (medium) or 5 (high), depending on the inherent contamination risk and the standard of equipment of the respective UNIT.

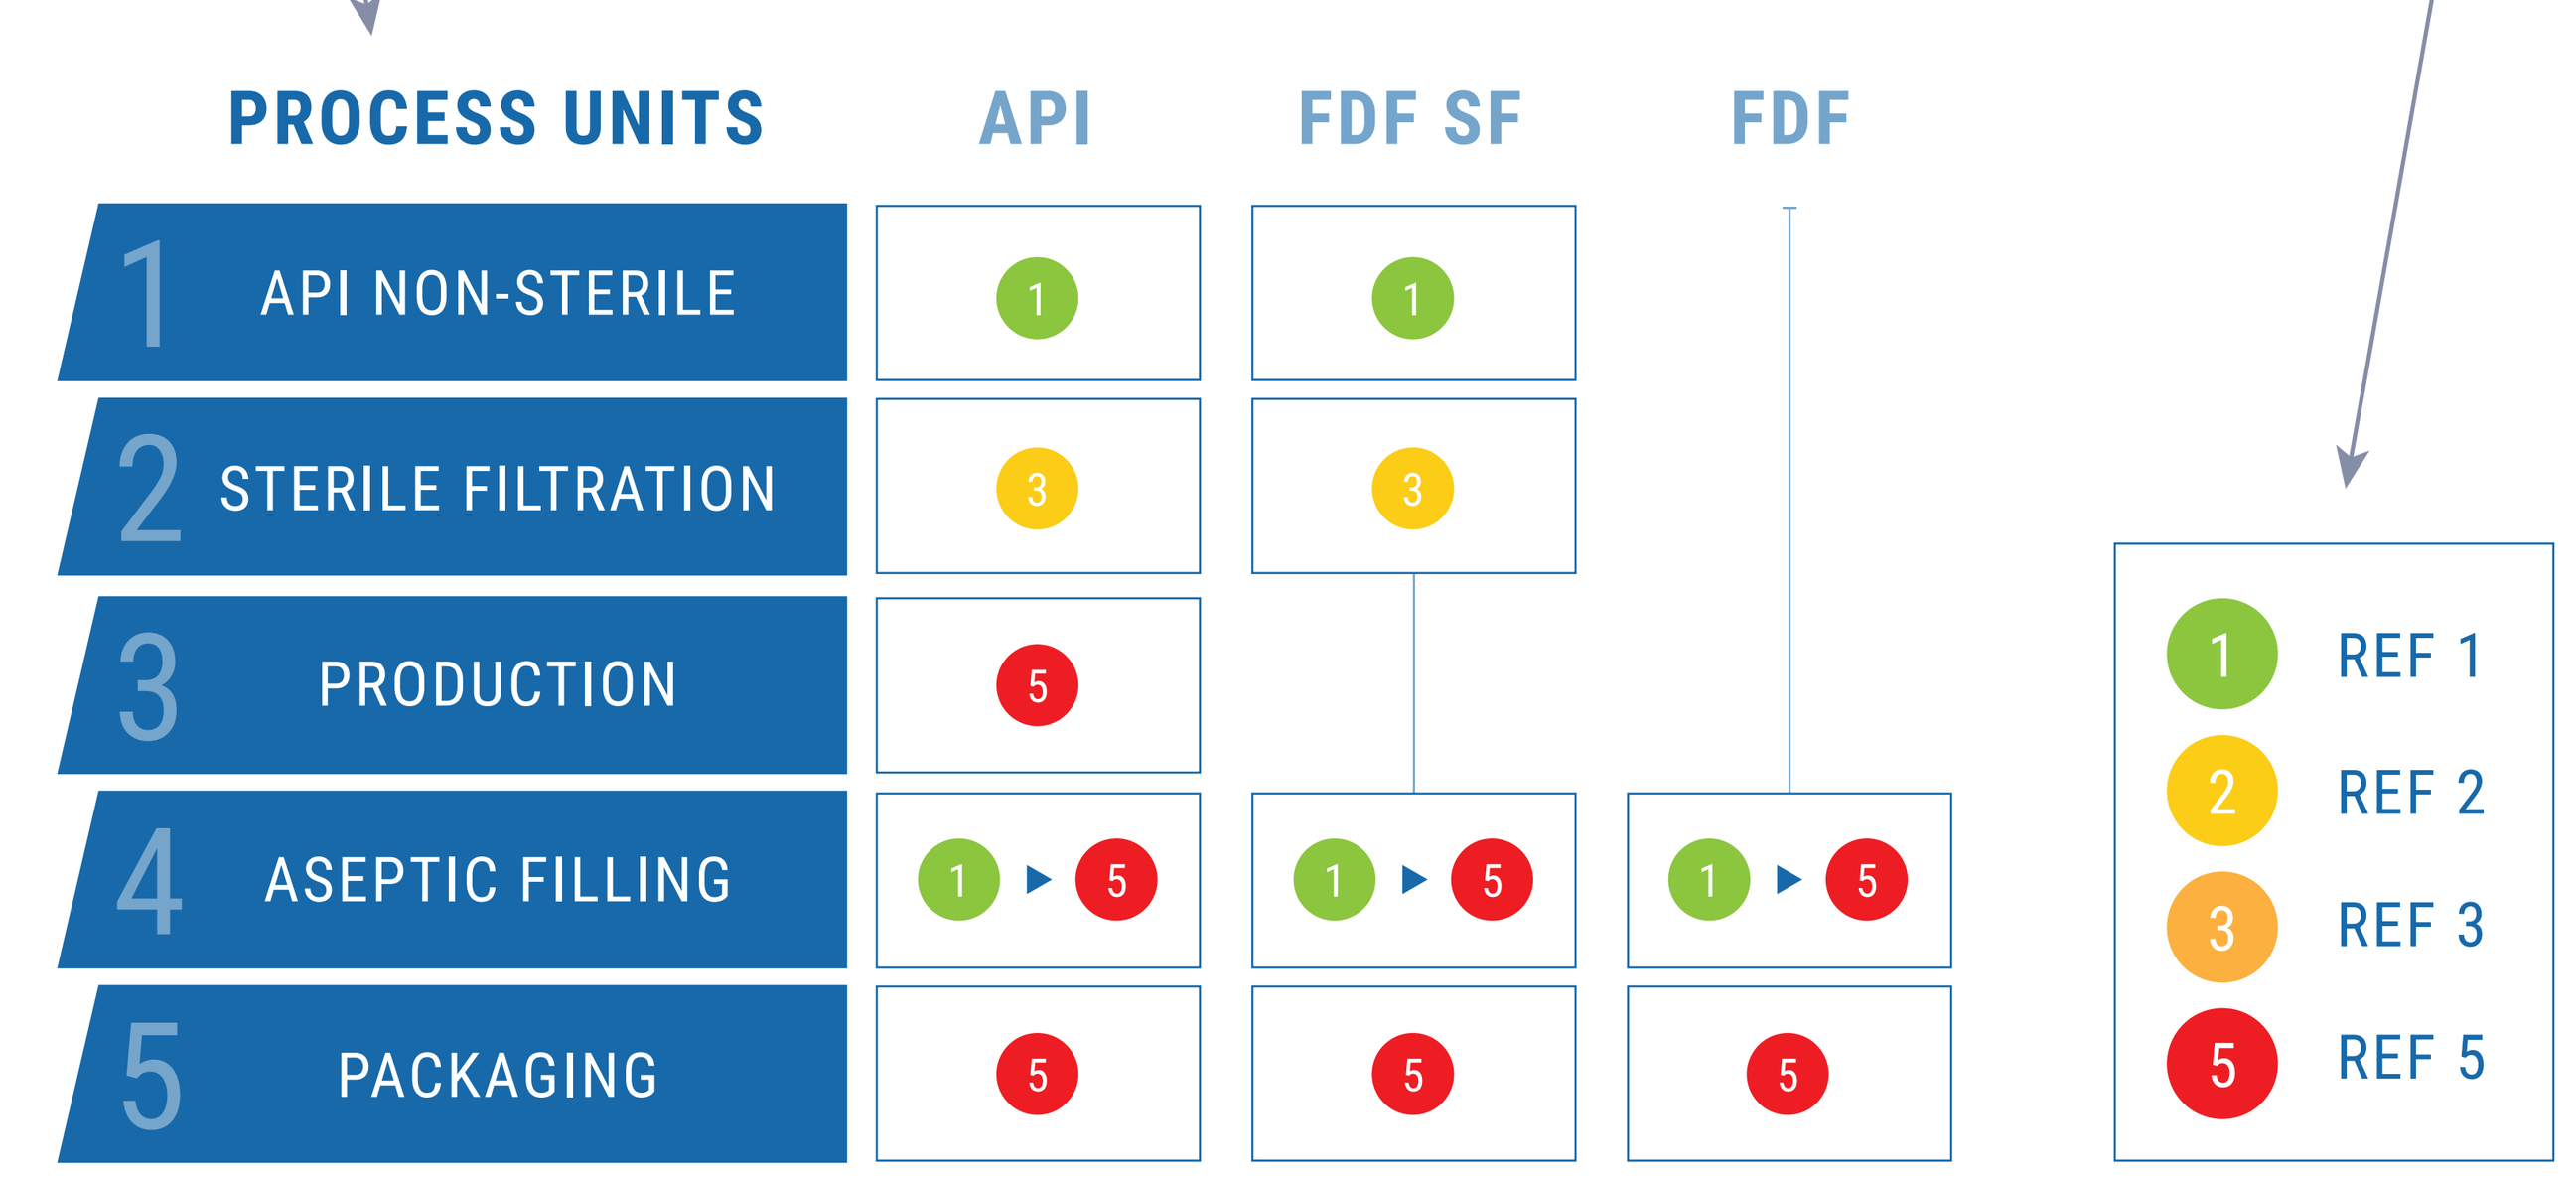

PROCESS UNITS
API
FDF SF
FDF
1 API NON-STERILE
2 STERILE FILTRATION
3 PRODUCTION
4 ASEPTIC FILLING
5 PACKAGING
REF 1
REF 2
REF 3
REF 5

Figure 1: Schematic overview of process units and related Risk Emphasis Factors (REFs)

Scoring between: 1 (excellent) to 5 (very poor or missing) or 100 (tremendous impact on overall sterility). The sum of all answered question scores from one UNIT is averaged to give the **UNIT AVERAGE RISK FACTOR** (the smaller, the lower the evaluated risk to the process with regard to the quality of its sterile product).

Each UNIT AVERAGE RISK FACTOR is multiplied by its corresponding unit REF to achieve the **UNIT RISK FACTOR**:

Unit Risk Factor = Unit Average Risk Factor x Unit REF

The HAZOP analysis is finally concluded by calculating the **TOTAL RISK FACTOR (TRF)**, which is the sum of all UNIT RISK FACTORs:

TRF = ∑ Unit Risk Factors

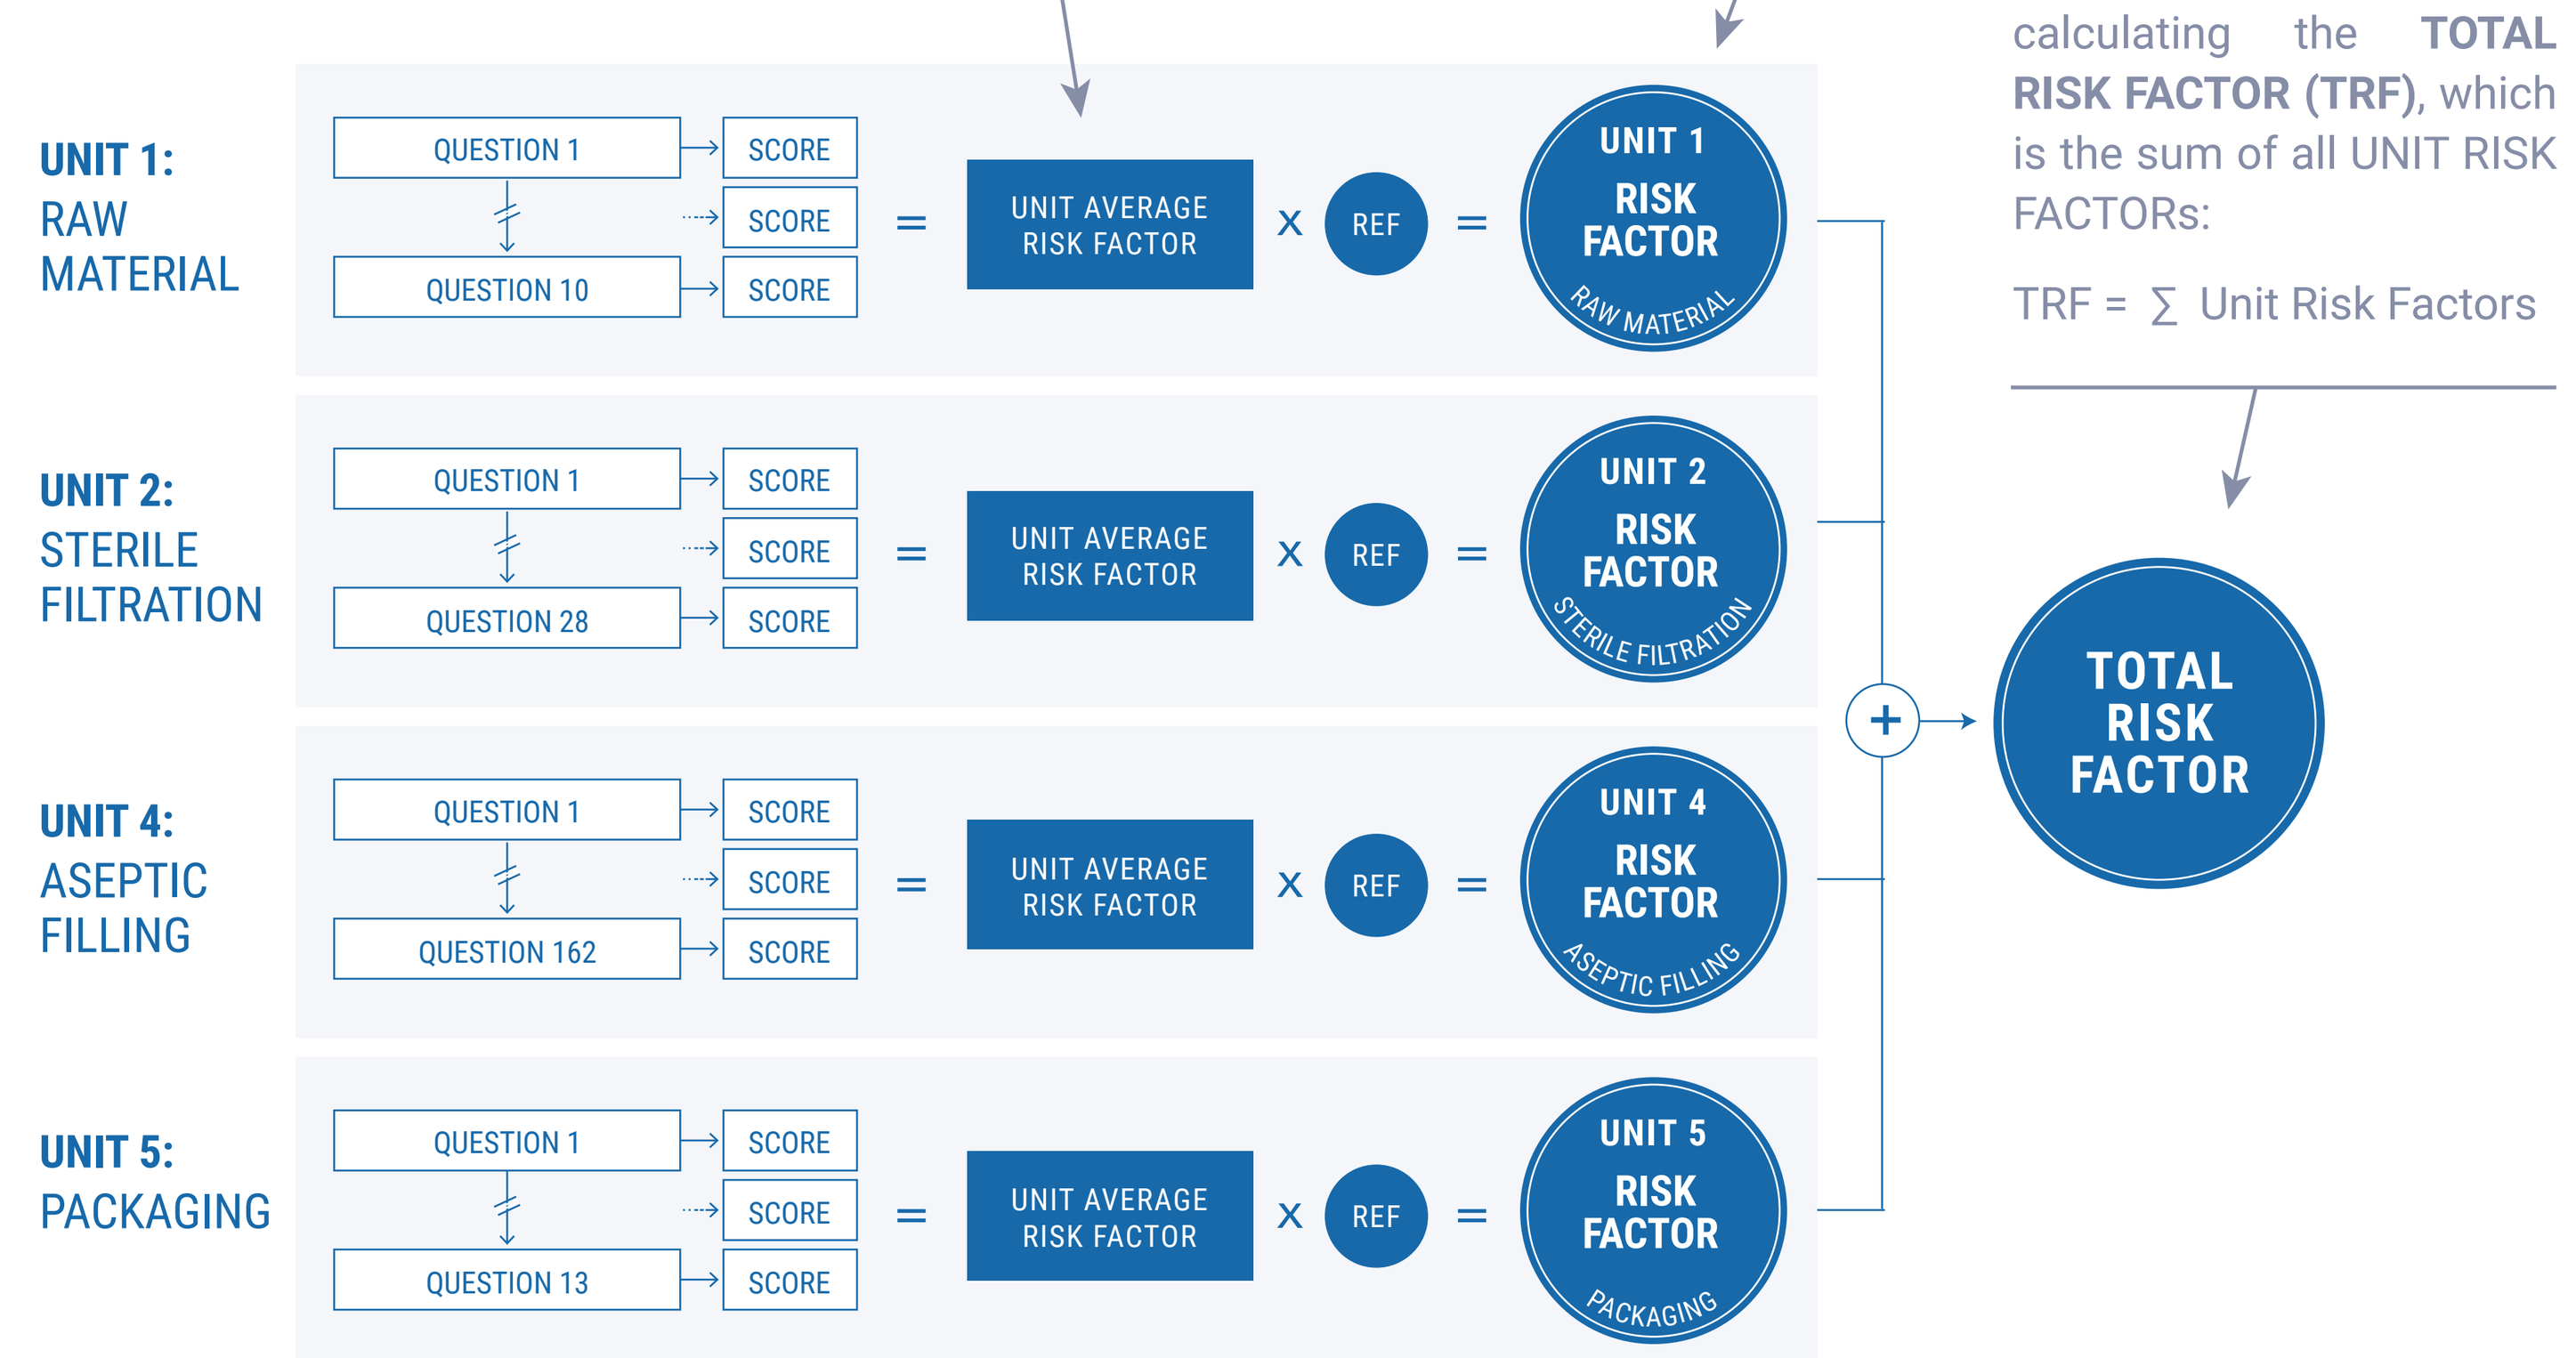

UNIT 1: RAW MATERIAL
UNIT 2: STERILE FILTRATION
UNIT 4: ASEPTIC FILLING
UNIT 5: PACKAGING
QUESTION 1
QUESTION 10
QUESTION 28
QUESTION 162
QUESTION 13
SCORE
UNIT AVERAGE RISK FACTOR
REF
UNIT 1 RISK FACTOR RAW MATERIAL
UNIT 2 RISK FACTOR STERILE FILTRATION
UNIT 4 RISK FACTOR ASEPTIC FILLING
UNIT 5 RISK FACTOR PACKAGING
TOTAL RISK FACTOR

Figure 2: Schematic overview of the Gapp Quality sterile product compliance risk analysis tool

The TRF provides definitive information about the overall risk of microbial contamination (sterility/endotoxins) for all production steps of an aseptic processing operation and allows the user to estimate the compliance status, as well as possible observations of future regulatory audits of the sterile plant.

Gapp Quality SPCRA Tool – Amendments

1 Variable Unit REFs

Variable REFs reflect the standard of equipment and type of filling technology used in "Aseptic Filling Units". This makes it possible to reward advanced aseptic technologies (e.g. isolators) that have a lower inherent contamination risk. Thus, an advanced filling line has a significantly lower TRF than a conventional filling line.
For example, for the sterile liquid filling line with sterile filtration (including lyophilization), the following REFs have been defined:

REF:	1	2	3	5
RAW MATERIAL UNIT:	X			
STERILE FILTRATION UNIT:			X	
ASEPTIC FILLING UNIT:	X	X	X	X
Closed isolator	X			
Isolator with "mouseholes"		X		
RABS* filling			X	
Conventional, open filling line				X
PACKAGING UNIT:			X	

** Restricted Access Barrier Systems*

2 Knock-Out Questions

Certain questions have a disproportionately high impact on overall sterility assurance. They represent deficiencies that could lead to product non-sterility or an FDA form 483 finding. To ensure that such risks are flagged and appropriately penalized, a score of 100 is assigned to a negative answer. These questions are referred to as "knock-out questions" (KO-questions). A negative answer may push the TRF from LOW to at least MODERATE, which should be considered unacceptable for a production plant.

- Prevent a severe risk factor being diluted when averaging all unit questions.
- Illuminate critical parameters that allow effective measures to be set and to get the senior management's attention if resources or investments are required.
- In a well-controlled sterile manufacturing plant a negative answer to a KO-question does not however necessitate the plant to be shut down, but defined CAPAs should be implemented with the highest priority.

Gapp Quality SPCRA Tool – Targets

- The final TRF should be in a "green range" (score 10-19,99) for FDF SF.
- No "KO-Question" should be answered with "100".
- Identified scored 100-CAPAS have the highest priority for remediation measures.

Real World Experience – Gapp Quality SPCRA Tool

Case #1 / Minimizing risk from MODERATE to LOW within one year

- European client / aseptic filling

2015 risk assessment: MODERATE risk for product non-sterility and regulatory non-compliance (TRF = 20.1)

Based on the risk assessment the following improvements were implemented:

QA/QC IMPROVEMENTS

- Increased training activities of staff, including preparation and usage of self-taped training videos
- QA-supervision in production during aseptic operations
- Execution of several microbiological studies, e.g. to support the holding time study of water samples, inclusion of bacterial spores included in the disinfectant effectiveness studies…
- Revision of the environmental monitoring (EM) program: usage of sterile/gamma-irradiated plates only for grade A/B, revised alert levels based on statistical trends, new trend analysis, justification of sample locations by FMEA risk assessment, active air monitoring is now going to be executed during filling and not just after filling (by using a new and less invasive remote active air monitoring system device)
- Investigation reports: strict usage of FMEA for batch disposition decisions

PRODUCTION/ TECHNICAL DEPARTMENT IMPROVEMENTS

- Transportation through the grade B corridor was resolved by double-bagging
- Simplification of filling line set-up (pre-assembled filling equipment, connection of tubes)
- Implemented changes in gowning: facemask is worn underneath the hood; 2 sterile operator gloves are now worn in grade A/B (before only one cotton glove was worn underneath the sterile glove)
- Media fills interventions now supported on a rationale and FMEA risk assessment

2016 risk assessment: LOW risk for product non-sterility and regulatory non-compliance (TRF = 17.7)

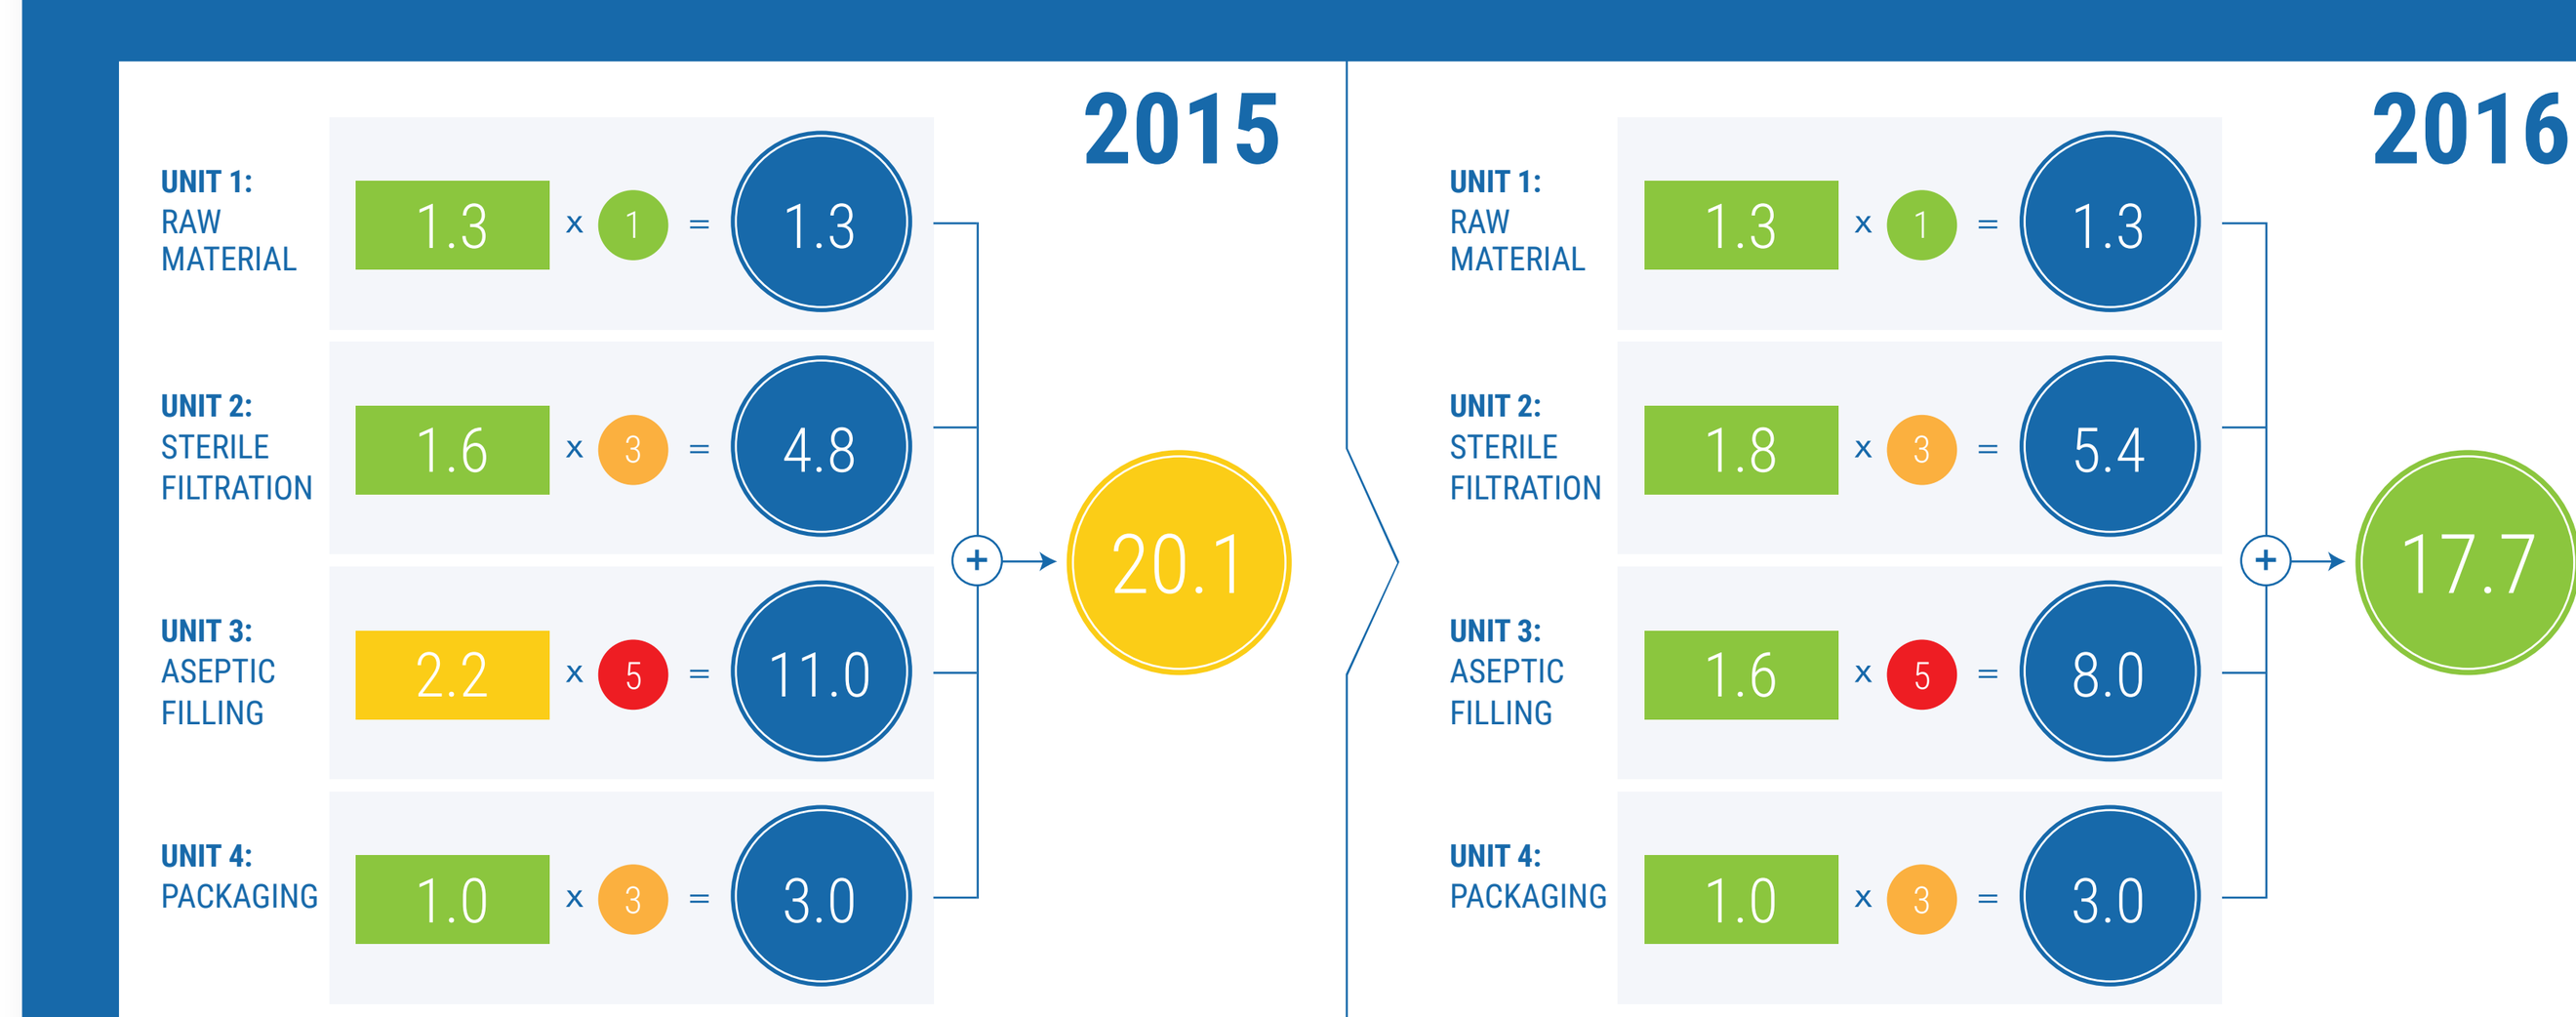

2015
UNIT 1: RAW MATERIAL 1.3 x 1 = 1.3
UNIT 2: STERILE FILTRATION 1.6 x 3 = 4.8
UNIT 3: ASEPTIC FILLING 2.2 x 5 = 11.0
UNIT 4: PACKAGING 1.0 x 3 = 3.0
20.1
2016
UNIT 1: RAW MATERIAL 1.3 x 1 = 1.3
UNIT 2: STERILE FILTRATION 1.8 x 3 = 5.4
UNIT 3: ASEPTIC FILLING 1.6 x 5 = 8.0
UNIT 4: PACKAGING 1.0 x 3 = 3.0
17.7

Case #2 / Assuring sterility compliance when faced with an FDA warning letter

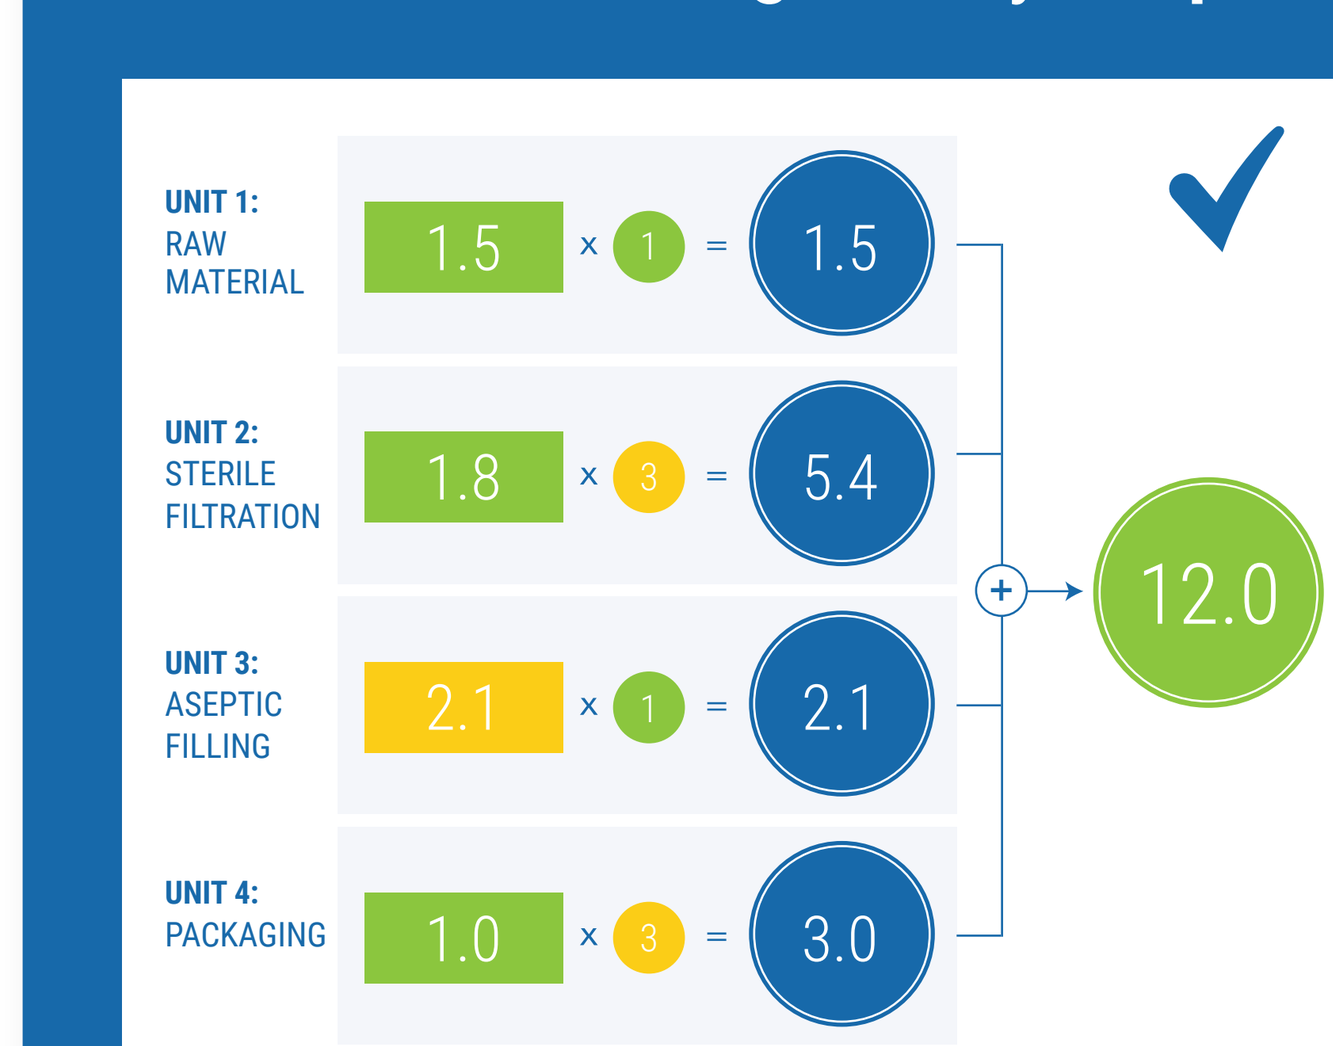

UNIT 1: RAW MATERIAL 1.5 x 1 = 1.5
UNIT 2: STERILE FILTRATION 1.8 x 3 = 5.4
UNIT 3: ASEPTIC FILLING 2.1 x 1 = 2.1
UNIT 4: PACKAGING 1.0 x 3 = 3.0
12.0

- Client with isolator filling cited with warning letter
- Variable REF was able to emphasize the impact of isolator *versus* open filling on product sterility
- **Due to isolator filling:**
 - REF = 1
 - Resulting risk is LOW for product non-sterility and regulatory non-compliance (TRF = 12.0)
- Independent SPCRA was sent to FDA and assured sterility compliance of client
- **If the unit had conventional open filling:**
 - REF = 5
 - Resulting risk would be MODERATE (TRF = 20.4)

Scope and Objective of SPCRA Tools

- SPCRA serves as investigative tool in case of deviations and as proactive tool for the prevention of non-compliance during product quality and regulatory audits
- Objective: to assess risk for product contamination
- KO-Questions are required! They may require production stoppage and/or assessment of product on the market, e.g. field alerts – but may „only" represent a compliance risk
- 36 Questions (Gappquality SPCRA Tool version 2017) are considered "Knock-out" due to high risk of product contamination
- Rating/Scoring should be done by an independent expert / 3rd party!
- In case of good outcome: very valuable tool to prove and improve process and product quality (e.g. 3rd party SPCRA for warning letter cited sites)
- SPCRA should be used proactively, and not to identify the root cause of quality issues

Risk Assessment Do's

- Successful quality management / risk assessment requires a skilled and experienced multifunctional team
- Support and high commitment to quality by the senior management
- Provide resources, invest in advanced technologies
- Commitment to honesty during execution of SPCRA
- Strengthen teamwork, support multidisciplinary cooperation
- Provide enough time (1-2 weeks) for risk assessment procedures

Summary sheet of SPCRA Tool (Case #2)

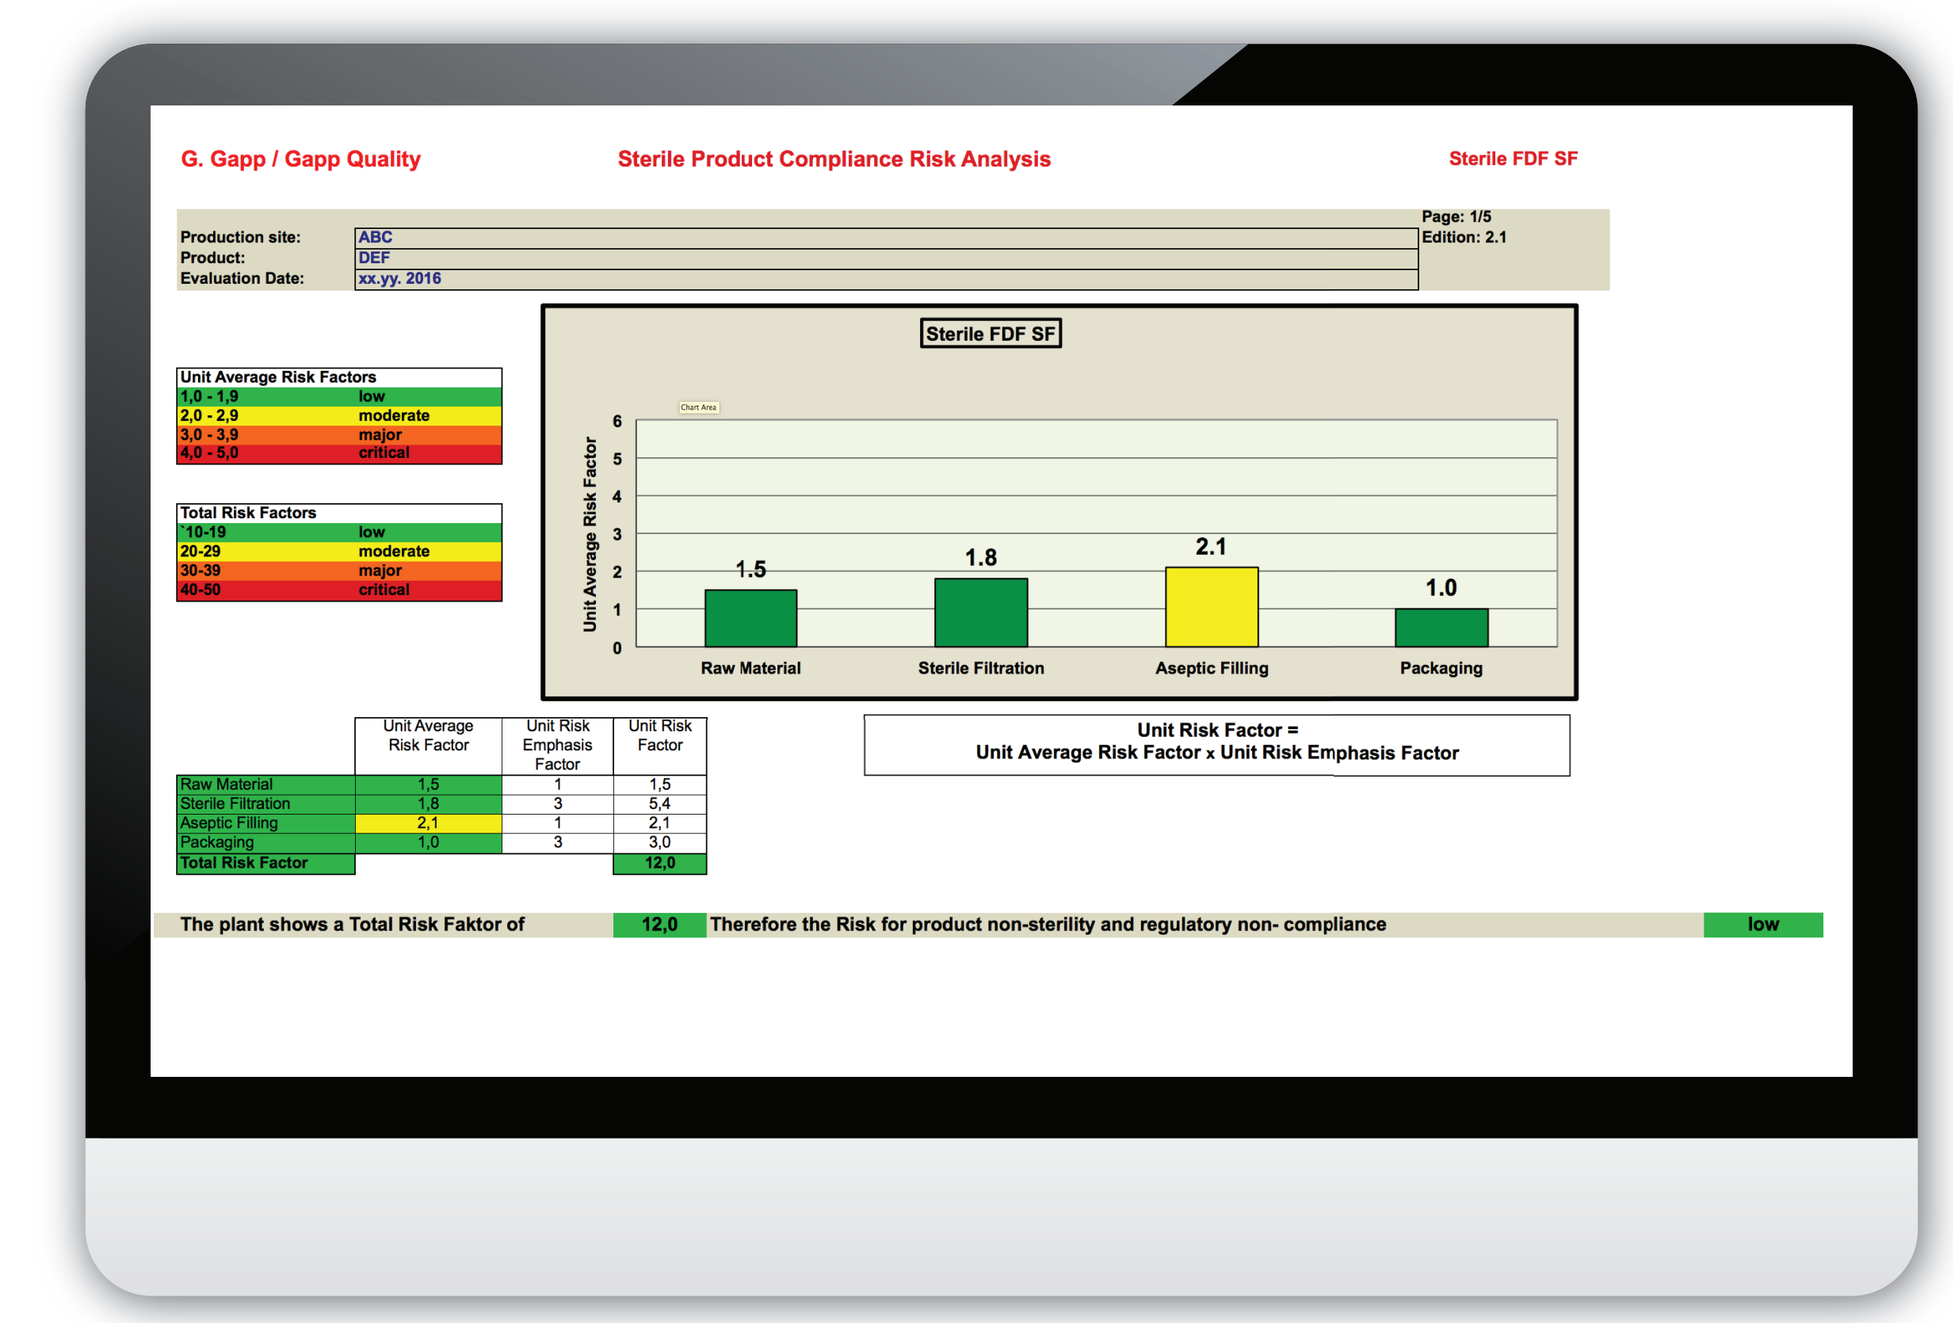

G. Gapp / Gapp Quality
Sterile Product Compliance Risk Analysis
Sterile FDF SF
Production site: ABC
Product: DEF
Evaluation Date: xx.yy. 2016
Page: 1/5
Edition: 2.1
Unit Average Risk Factors
Total Risk Factors
Sterile FDF SF
Unit Average Risk Factor
Raw Material
Sterile Filtration
Aseptic Filling
Packaging
Unit Risk Factor = Unit Average Risk Factor x Unit Risk Emphasis Factor
The plant shows a Total Risk Faktor of
12,0
Therefore the Risk for product non-sterility and regulatory non- compliance
low

References

[1] U.S. Food and Drug Administration Department of Health and Human Services: Pharmaceutical cGMPS for the 21st Century – A Risk Based Approach. Final Report – Fall 2004 [Internet]

[2] International Conference on Harmonisation of Technical Requirements for Registration of Pharmaceuticals for Human Use: ICH Tripartite Guideline, Quality Risk Management Q9. Current Step 4 version dated 9 November 2005 [Internet]

[3] International Conference on Harmonisation of Technical Requirements for Registration of Pharmaceuticals for Human Use: ICH Tripartite Guideline, Pharmaceutical Quality System Q10. Current Step 4 version dated 4 June 2008 [Internet]

[4] ISO 14971:2007 - Application of risk management to medical devices [Internet]

[5] EudraLex – Volume 4 –Guidelines for Good Manufacturing Practice for Medicinal Products for Human and Veterinary Use. Part 1, Chapter 1: Pharmaceutical Quality System [Internet]

[6] Gapp G, Holzknecht P. Risk analysis of sterile production plants: a new and simple, workable approach. PDA J Pharm Sci Technol. 2011 May-Jun;65(3):217-26

Acknowledgements

SCIENCE WRITING ULRIKE POSCH

Thank you to SCIENCEWRITING ULRIKE POSCH for editorial support, layout and design. **www.sciencewriting.at**

2017 PDA ANNUAL MEETING | April 3-5 | Anaheim, CA